Cell-derived vesicles: Unraveling the science of novel vesicles with therapeutic promises

Hui-Chong Lau[1], Jihye Lee[1], Dong Woo Han[1], Jinhee Park[1], Soyeon Kim[1] Jae Young Kim[1], Suhee Kim[2], Hye-Jung Kim[2], and Seung Wook Oh[1]*
[1] MDimune Research Institute, MDimune Inc., Seoul, Republic of Korea
[2]New Drug Development Center, KBIO Osong Medical Innovation Foundation, Cheongju-si, Chungbuk, Republic of Korea
*Correspondence: swoh@mdimune.com

EXTRUSION TECHNOLOGY

Cell-derived vesicles (CDVs) are nanosized vesicles produced by serially extruding cells through small pores. A growing number of studies have implicated their therapeutic potentials, with superior yield compared to other extracellular vesicles (EVs). However, two key objectives remain to be accomplished to fully demonstrate the utility of CDVs in clinical applications. First, the manufacturing process has to be developed to allow a large-scale production of CDVs. Next, these novel vesicles need to be thoroughly characterized at multiple levels to establish the best therapeutic strategy for CDVs. Here, we developed a scalable manufacturing process for CDVs and further characterized these vesicles to meet the objectives of therapeutics applications.

PRODUCTION PROCESS, YIELD, MEASURENMENT RANGE & CHARACTERIZATION

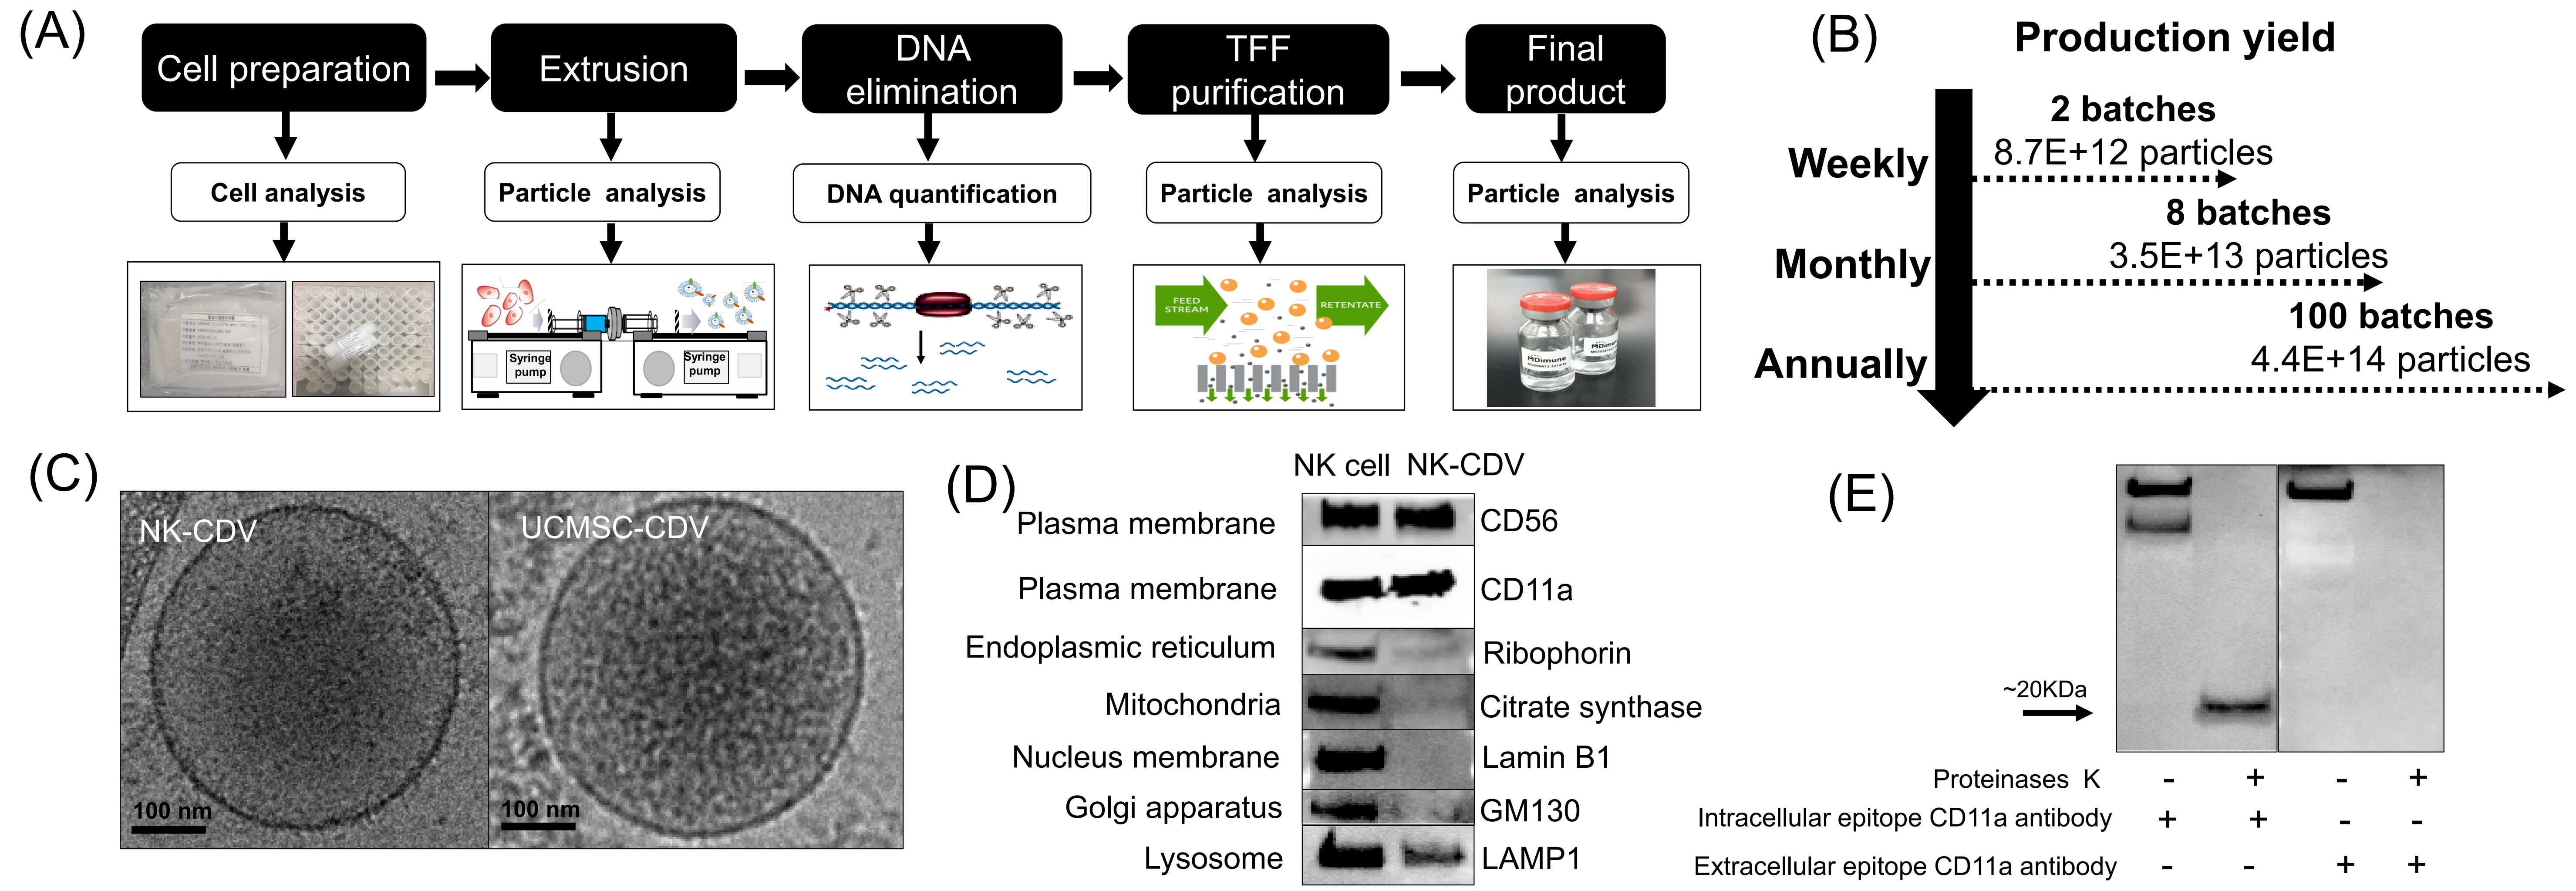
(A)
Cell preparation
Extrusion
DNA elimination
TFF purification
Final product
Cell analysis
Particle analysis
DNA quantification
Particle analysis
Particle analysis
Syringe pump
FEED STREAM
RETENTATE
(B)
Production yield
Weekly
2 batches
8.7E+12 particles
Monthly
8 batches
3.5E+13 particles
Annually
100 batches
4.4E+14 particles
(C)
NK-CDV
UCMSC-CDV
100 nm
(D)
NK cell NK-CDV
Plasma membrane CD56
Plasma membrane CD11a
Endoplasmic reticulum Ribophorin
Mitochondria Citrate synthase
Nucleus membrane Lamin B1
Golgi apparatus GM130
Lysosome LAMP1
(E)
~20KDa
Proteinases K - + - +
Intracellular epitope CD11a antibody + + - -
Extracellular epitope CD11a antibody - - + +

Figure 1: (A) Manufacturing-scale production process of CDVs developed at MDimune. (B) Current production capacity and yield of USMSC-CDVs on a weekly, monthly, or annual basis. More than 4E+14 particles produced a year can support various pre-clinical and early clinical stages, which can be further upgraded to meet the larger clinical requirement. (C) Representative images of CDVs examined using cryo-TEM. CDV has a spherical shape with a lipid bilayer, similar to other EVs. (D) Expression of the plasma membrane and subcellular organelle membrane markers. CDVs are mainly composed of the plasma membrane proteins, while containing the diminished amount of subcellular organelle membrane proteins. (E) The topology of CDV membrane. CDVs were treated with proteinases K to digest surface portion of membrane proteins. Anti-CD11a antibody with intracellular epitopes detects the intra-vesicular fragment of CD11a after proteinases K digestion, whereas the antibody with extracellular epitope does not. This result suggests that extruded CDV maintains the membrane topology of the cell.

Table 1: QC parameter and current measurement range of CDVs

Category	Parameter	Instrument/method	Current measurement range	
			NK-CDV	UCMSC-CDV
Purified CDV	Size (nm)	DLS	160~200	135-175
	PDI	DLS	0.1~0.3	0.12-0.42
	Particles/batch	NTA	~5E+12 particles/1E+9	~4E+12 particles/1E+8
	Extracted DNA (ng/batch)	Pico green dsDNA assay kit	≤10	≤10
	Benzonase (Unit/batch)	ELISA	≤20	≤30
	Surface marker	FACS (Aldehyde sulfate bead)	>70% CD11a, CD56	>70% CD63, CD73, CD90, CD29

Physical and biochemical properties of CDVs were characterized to control the quality of CDVs at each batch of production. Current measurement range was established based on the results from multiple batches of production.

MULTI-OMICS STUDY

Multi-omics profiling was carried out with three independent batches of NK cells and NK-CDVs to comprehend the fundamental basis of the molecular contents within CDVs.

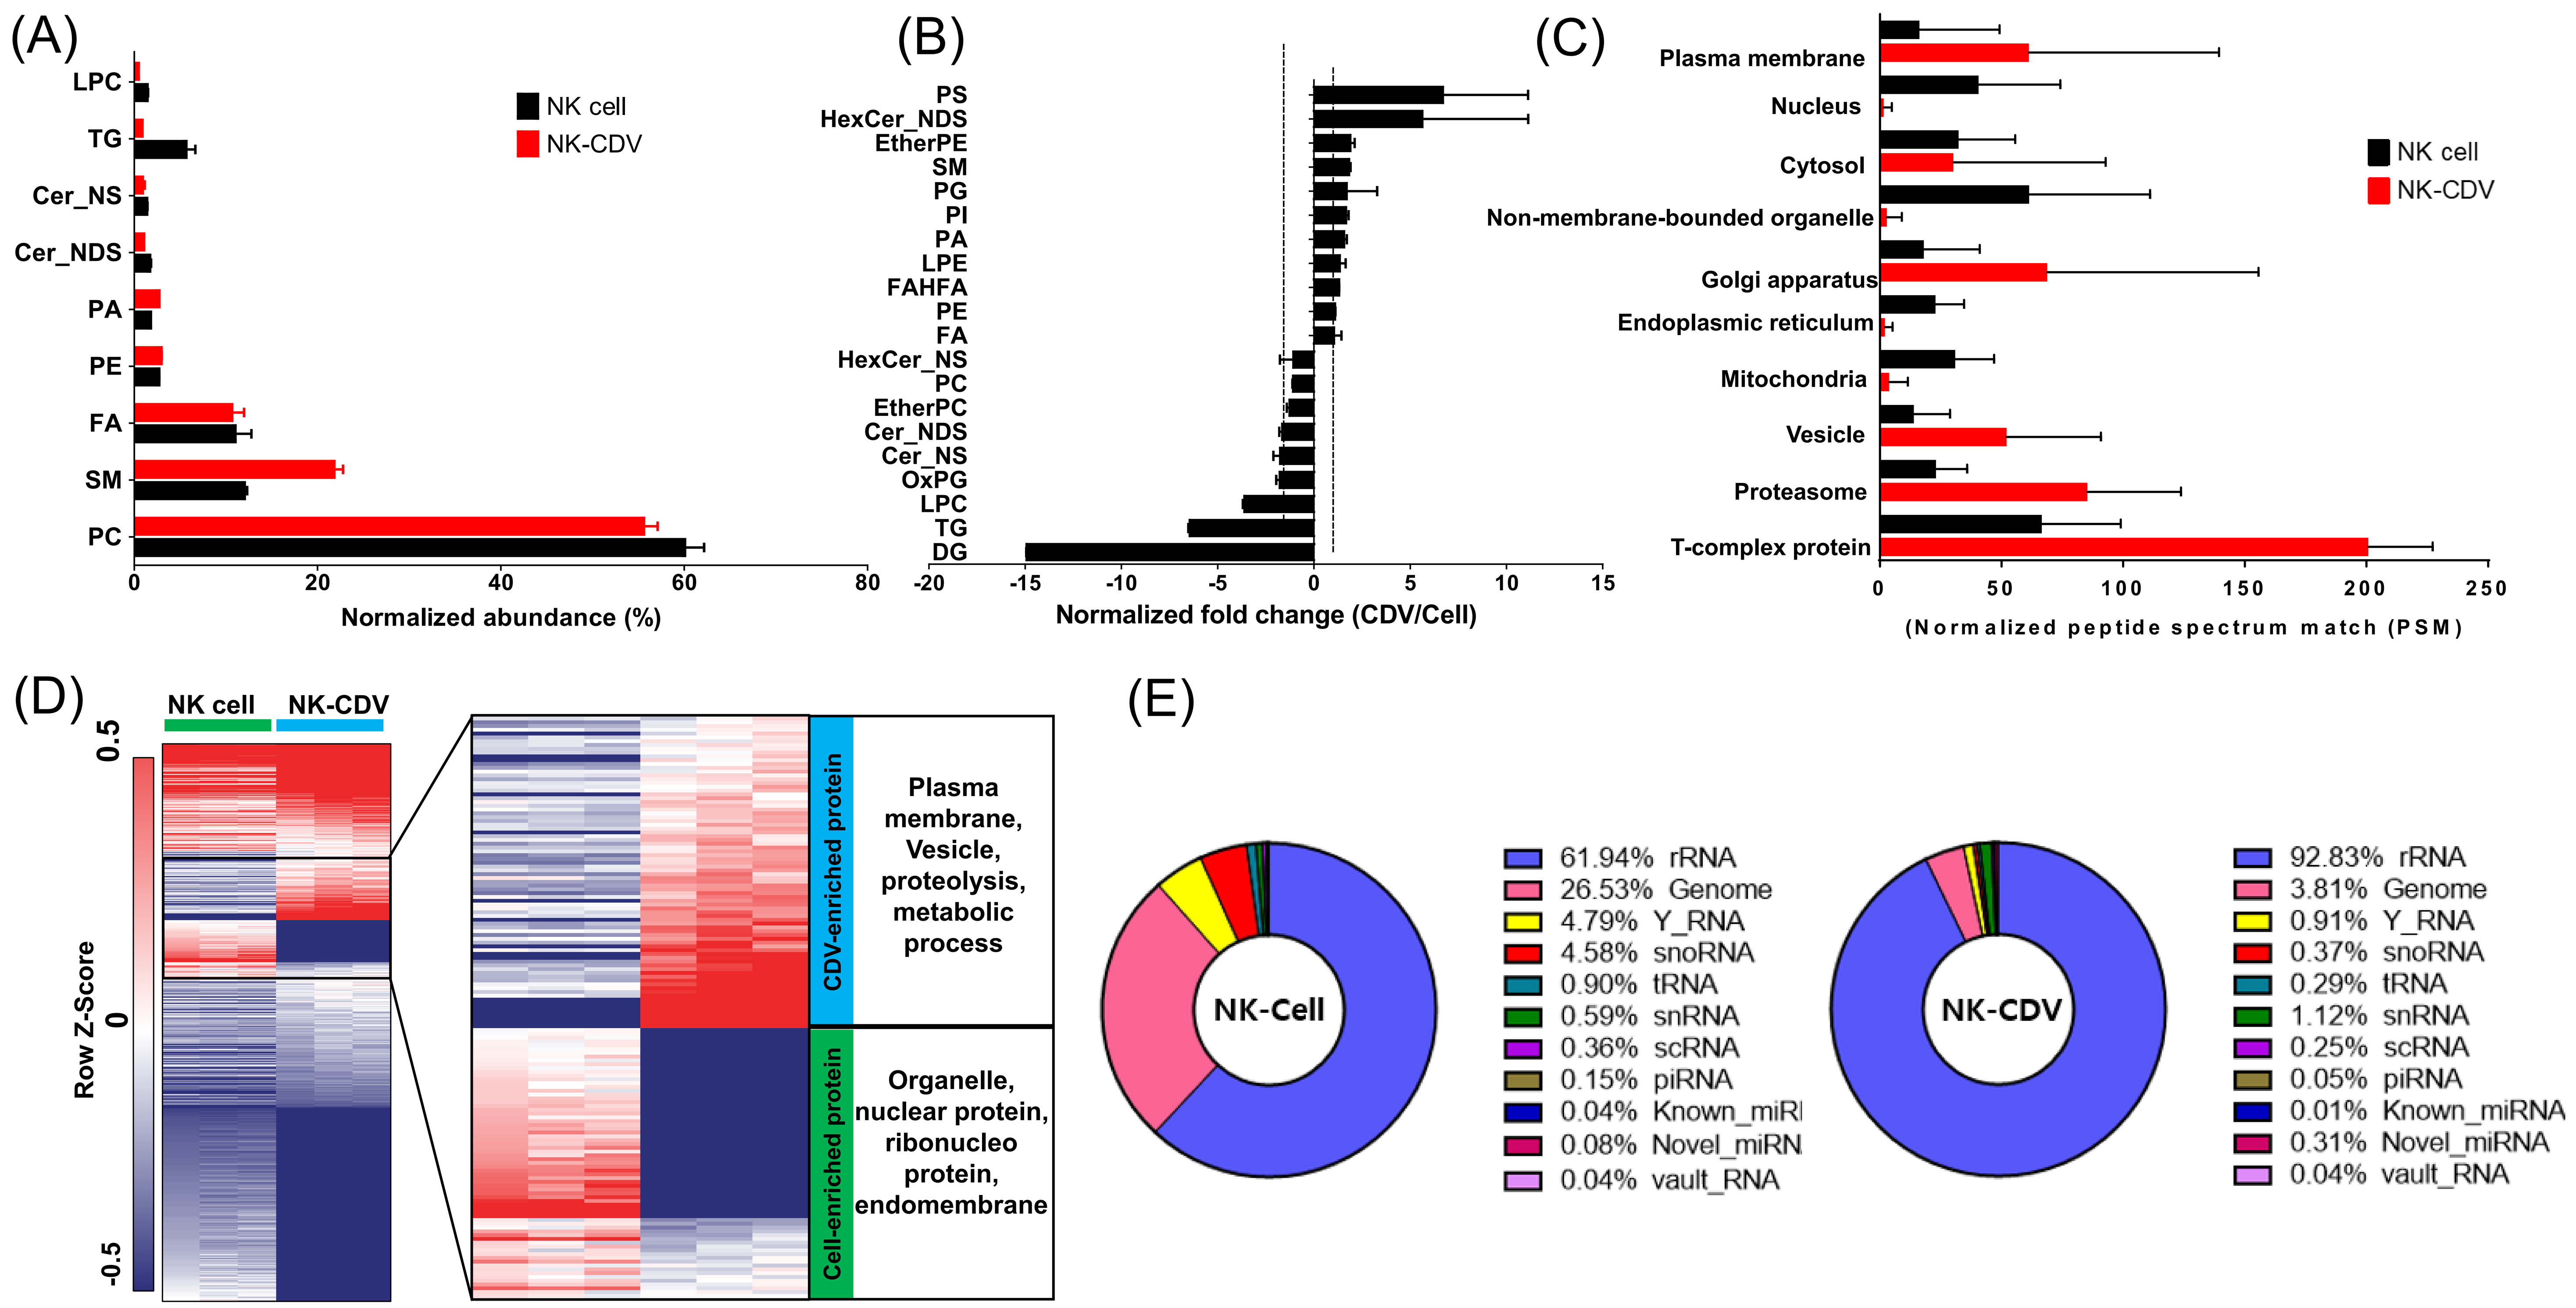
(A)
LPC
TG
Cer_NS
Cer_NDS
PA
PE
FA
SM
PC
NK cell
NK-CDV
0 20 40 60 80
Normalized abundance (%)
(B)
PS
HexCer_NDS
EtherPE
SM
PG
PI
PA
LPE
FAHFA
PE
FA
HexCer_NS
PC
EtherPC
Cer_NDS
Cer_NS
OxPG
LPC
TG
DG
-20 -15 -10 -5 0 5 10 15
Normalized fold change (CDV/Cell)
(C)
Plasma membrane
Nucleus
Cytosol
Non-membrane-bounded organelle
Golgi apparatus
Endoplasmic reticulum
Mitochondria
Vesicle
Proteasome
T-complex protein
NK cell
NK-CDV
0 50 100 150 200 250
(Normalized peptide spectrum match (PSM))
(D)
NK cell
NK-CDV
0.5
Row Z-Score
0
-0.5
CDV-enriched protein
Plasma membrane, Vesicle, proteolysis, metabolic process
Cell-enriched protein
Organelle, nuclear protein, ribonucleo protein, endomembrane
(E)
NK-Cell
61.94% rRNA
26.53% Genome
4.79% Y_RNA
4.58% snoRNA
0.90% tRNA
0.59% snRNA
0.36% scRNA
0.15% piRNA
0.04% Known_miRI
0.08% Novel_miRN
0.04% vault_RNA
NK-CDV
92.83% rRNA
3.81% Genome
0.91% Y_RNA
0.37% snoRNA
0.29% tRNA
1.12% snRNA
0.25% scRNA
0.05% piRNA
0.01% Known_miRNA
0.31% Novel_miRNA
0.04% vault_RNA

Figure 2: Multi-omics analysis of NK cells and NK-CDVs. (A) Distribution of lipid class in NK cells and NK-CDVs was based on the average normalized abundances. Phosphatidylcholine (PC) is the most abundant lipid class in both NK cells and NK-CDVs followed by sphingomyelin (SM) and fatty acid (FA). CDVs contains more SM than cells whereas TG (triacylglycerol) is much higher in cells than CDVs. (B) The enrichment of lipid class was based on the normalized fold change (CDV/Cell). (C) In proteomics, to meet the criteria of significance, all genes with fold change > 2 and p-value < 0.05 were used. Gene Ontology (GO) annotation analysis of the identified cellular components for NK cells and NK-CDVs were further grouped into 10 main categories. Based on the normalized PSM, plasma membrane proteins are enriched in NK-CDVs whereas the organelle membrane proteins are more abundant in cells. (D) Heat map analysis of GO annotated protein list showed a distinctive protein enrichment profile in CDVs and cells. CDVs are enriched with proteins related to the plasma membrane and proteolysis whereas organelles, nuclear-related proteins are enriched in cells. (E) The RNA composition profile showed that rRNA is the main RNA types in cells and NK-CDVs. MicroRNA has only about 0.04% and 0.013% in cells and CDVs, respectively.

CONCLUSIONS

- We developed a scalable manufacturing and quality control process for CDVs.
- Molecular and biochemical analyses show that the NK-CDV is enriched with the plasma membrane, whereas most subcellular organelles and non-membrane bound organelles are enriched in cells.
- NK cells and NK-CDV shared the similar lipid class and RNA profiles, except the genomic RNA is greatly reduced in CDV.
- Comprehensive characterization of CDVs reveals that CDVs are a powerful alternative to cells with conserved membrane properties.